Biological activity of FLX475, an oral CCR4 antagonist, as monotherapy and in combination with pembrolizumab in advanced cancer

Juraj Adamik*, Adam Grant*, Damian Trujillo, Rakesh Kumar Goyal, William Ho, Michael Chisamore, Shoji Ikeda, Marvin Au, Denise de Almeida Nagata, Eugene Lurie, Qingfeng Zhu, Robert A. Anders, Dirk G. Brockstedt, Mohsen Sabouri Ghomi, Paul D. Kassner; Merck & Co., Inc., Rahway NJ, USA; Johns Hopkins University School of Medicine, Baltimore MD, USA; RAPT Therapeutics, Inc., South San Francisco CA, USA

FLX475 is a potent and selective CCR4 antagonist, designed to block immunosuppressive regulatory T cell (Treg) migration into the tumor microenvironment (TME), which has the potential to overcome immune resistance and broaden clinical efficacy to a variety of conventional and immunotherapy-based approaches. In a recent interim clinical update from the ongoing FLX475-02 Phase 1/2 trial (NCT03674567), evidence of monotherapy and combination activity were reported. Here, we present biomarker data from patients with multiple tumor types treated with FLX475 as monotherapy and in combination with pembrolizumab. These data support the beneficial effects of FLX475 in modification of the TME and promotion of anti-cancer immunity.

As determined by flow cytometry, a small but significant increase in proportion of circulating Treg (CD4+CD25+CD127-/low) was observed in patients by day 8 of treatment. Immunohistochemistry (IHC) revealed that FLX475 monotherapy increased CD8/Foxp3 density ratio, increased distance between CD8+ and Foxp3+ cells, and reduced migration of Foxp3+ cells from stroma to viable tumor regions. RNA-seq data derived from tumor biopsies prior to (n = 33), and after approximately 6 weeks of treatment (n = 22, paired samples) with FLX475+/-pembrolizumab were compared to published biopsy data from anti-PD-(L)1 treated patients. Transcriptomic profiles from tumor biopsies of FLX475 monotherapy treated patients exhibited significant changes in immune pathways to resemble profiles from patients with favorable clinical outcome to anti-PD-(L)1 treatment. Analysis of paired biopsies from both FLX475/pembrolizumab and anti-PD-1 regimens showed significantly increased T cell signatures. However, FLX475/pembrolizumab prevented coordinated increase of Treg cell signatures observed in the TME of patients treated with anti-PD-1 alone. Consistent with this finding, increased expression of *CCR4* and its ligands *CCL17* and *CCL22* were observed in biopsies of patients receiving anti-PD-1 treatment but not FLX475/pembrolizumab. To identify patients more likely to benefit from FLX475/pembrolizumab therapy, baseline transcriptomic profiles were analyzed. Patients with clinical benefit (CR, PR, and stable disease ≥6 months) were found to have elevated T_{reg} populations compared to those without clinical benefit (PD and SD <6 months). This phenomenon was not observed in external anti-PD-1 datasets.

FLX475 monotherapy and in combination with pembrolizumab result in beneficial changes in the TME. FLX475 monotherapy appears to modify the TME toward a phenotype associated with response to anti-PD-(L)1. Baseline markers associated with favorable outcome are different for the combination treatment compared to anti-PD-1 monotherapy suggesting that patients with enriched intratumoral Tregs might benefit from FLX475/pembrolizumab combination than pembrolizumab alone.

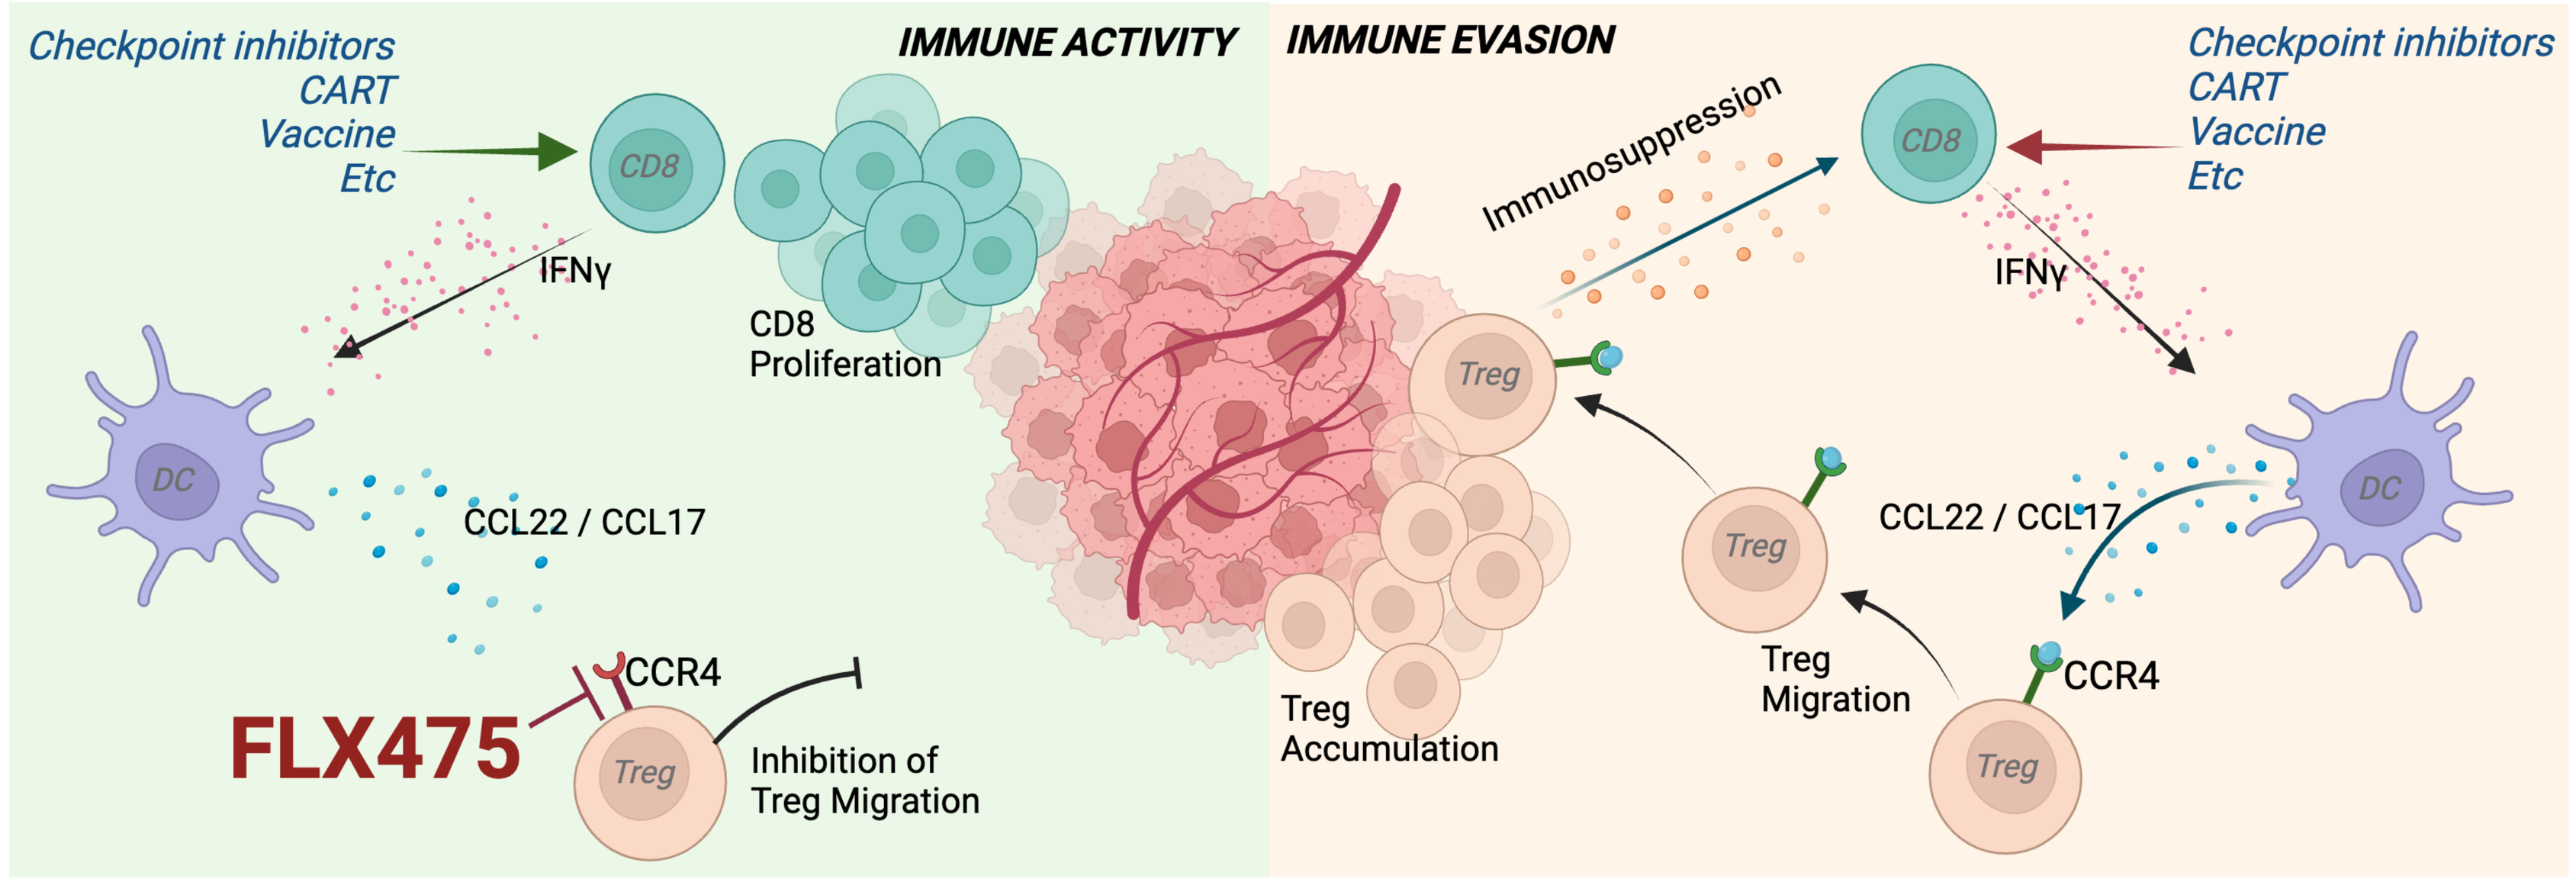
Checkpoint inhibitors
CART
Vaccine
Etc
IMMUNE ACTIVITY
IMMUNE EVASION
Immunosuppression
CD8
IFNγ
CD8
Proliferation
DC
CCL22 / CCL17
CCR4
FLX475
Treg
Inhibition of
Treg Migration
Treg
Accumulation
Treg
Migration
CCR4

- *Proposed mechanism of action for FLX475 blocking CCR4-mediated migration of Tregs into the TME and promoting anti-cancer immunity.*

FLX475 monotherapy alters TME expression to resemble anti-PD-(L)1 responders

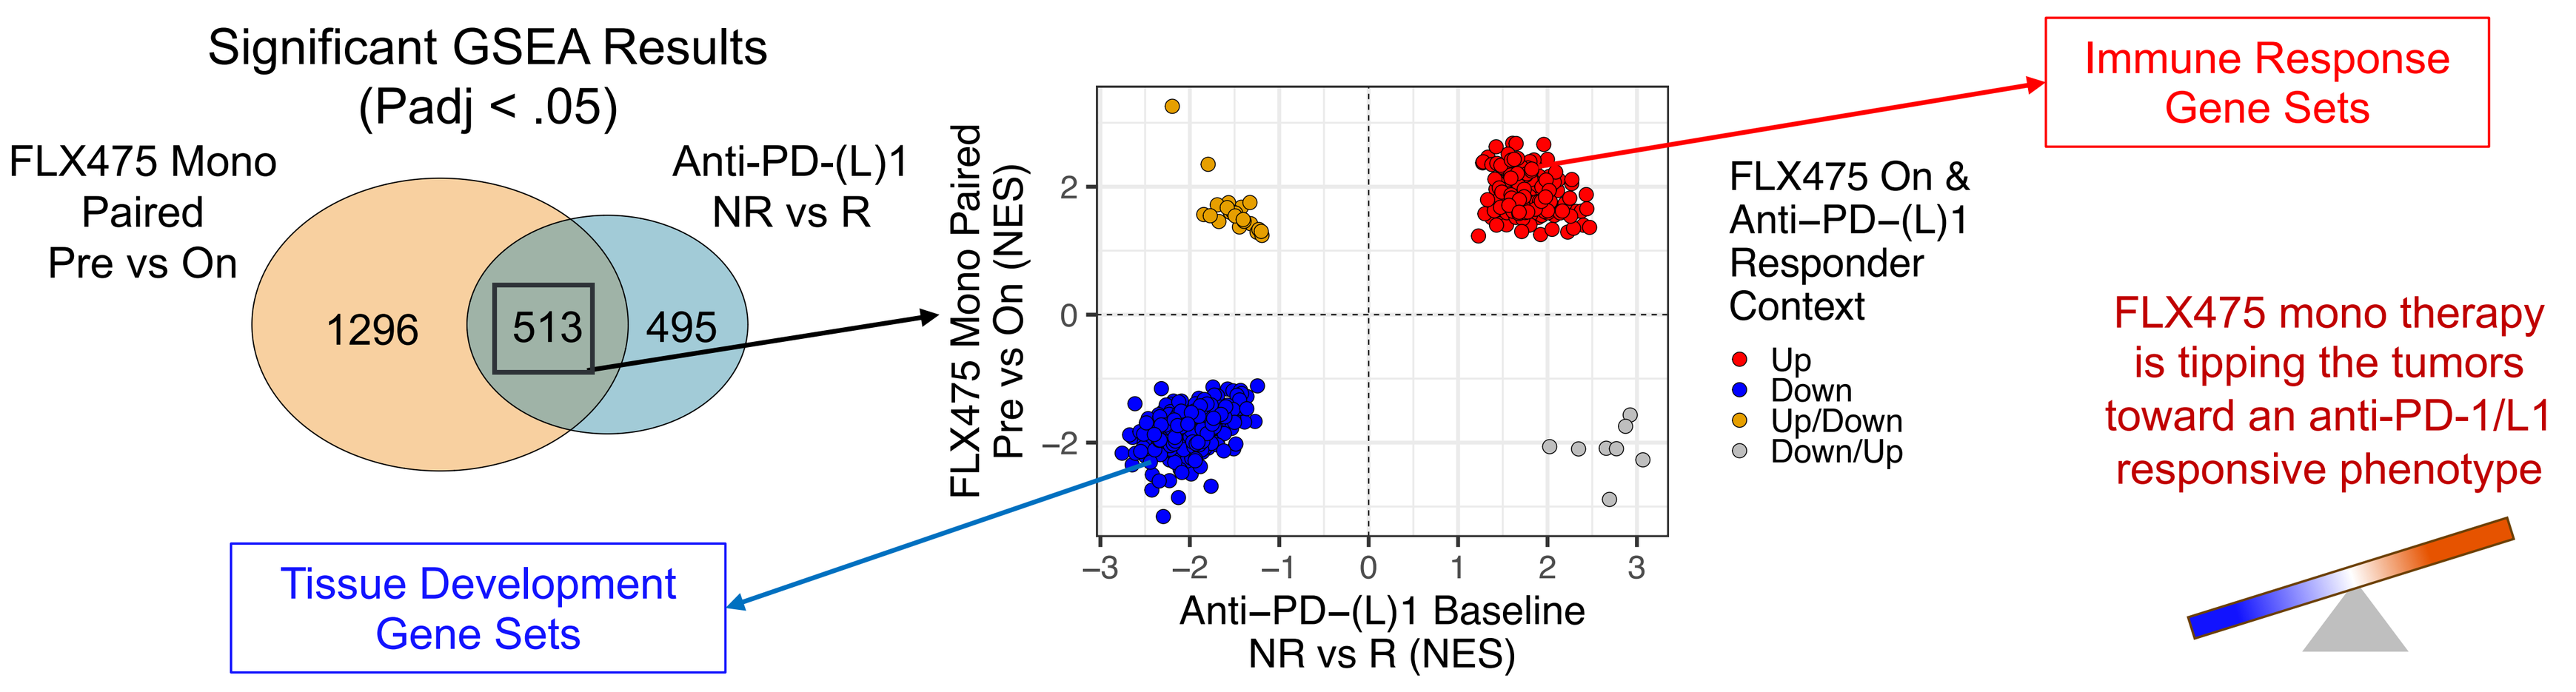
Significant GSEA Results
(Padj < .05)
FLX475 Mono
Paired
Pre vs On
Anti-PD-(L)1
NR vs R
1296
513
495
Tissue Development
Gene Sets
FLX475 Mono Paired
Pre vs On (NES)
Anti−PD−(L)1 Baseline
NR vs R (NES)
FLX475 On &
Anti−PD−(L)1
Responder
Context
Up
Down
Up/Down
Down/Up
Immune Response
Gene Sets
FLX475 mono therapy
is tipping the tumors
toward an anti-PD-1/L1
responsive phenotype

- *GSEA: gene set enrichment analysis, NR: non-responder, R: responder, NES: normalized enrichment score from GSEA*
- *FLX475 increases expression of gene sets associated with immune response and decreases tissue development-associated gene signatures*
- *Anti-PD-1/PD-L1 Datasets obtained from references 1, 2, 4, 5*

FLX475 limits CCR4-associated Treg accumulation in the TME observed with anti-PD-1 treatment

Contrary to FLX475/pembrolizumab, anti-PD-1 monotherapy upregulates CCR4 and CCR4 ligands favoring Treg cell recruitment into the tumor

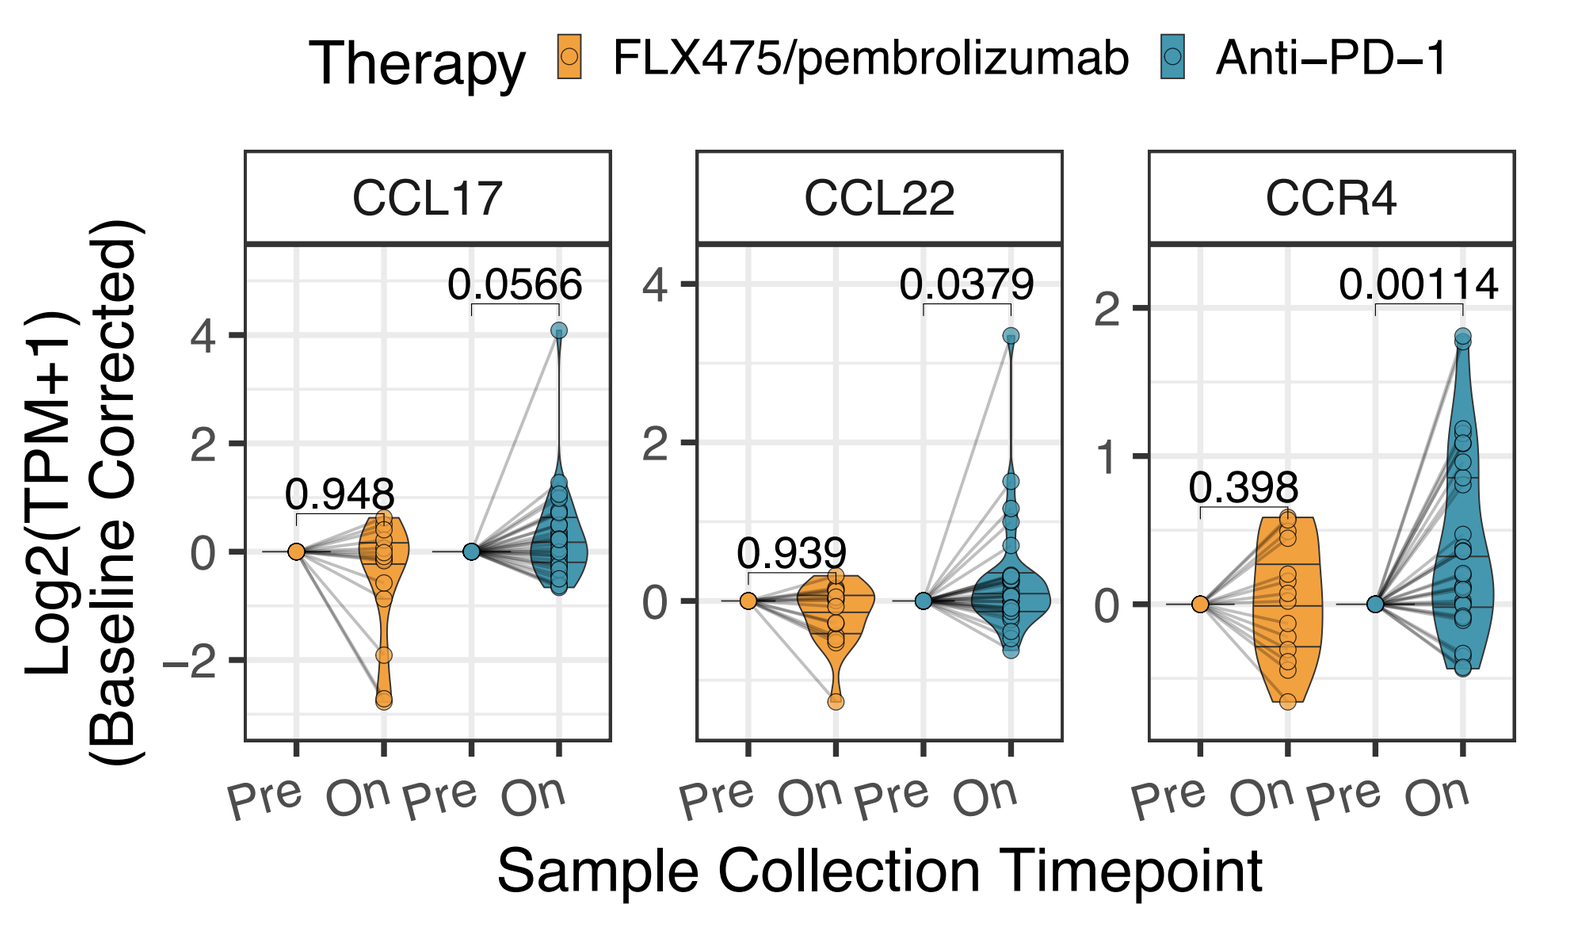
Therapy
FLX475/pembrolizumab
Anti−PD−1
CCL17
CCL22
CCR4
0.948
0.0566
0.939
0.0379
0.398
0.00114
Log2(TPM+1)
(Baseline Corrected)
Pre
On
Sample Collection Timepoint

- *CPI-naïve patient samples analyzed by RNAseq of turmor biopsy*
- *One-sided paired t-test was used to test significance*
- *Anti-PD-1 datasets obtained from references 2, 5*

FLX475/pembrolizumab results in CD8 increase without the accumulation of Tregs observed with anti-PD-1 monotherapy

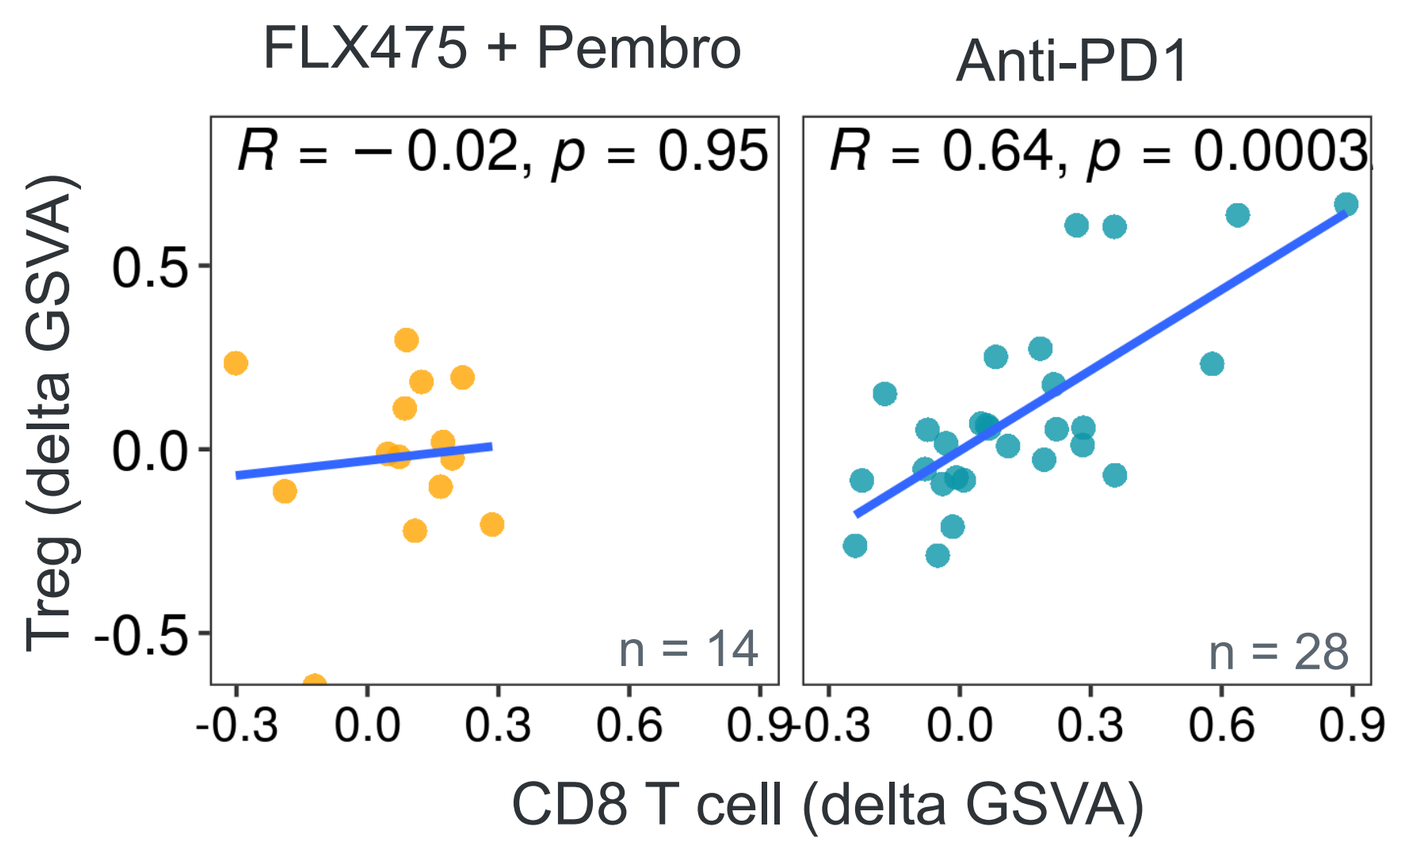
FLX475 + Pembro
Anti-PD1
R = −0.02, p = 0.95
R = 0.64, p = 0.0003
n = 14
n = 28
Treg (delta GSVA)
CD8 T cell (delta GSVA)

- *Gene Set Variance Analysis (GSVA) was used to evaluate immune infiltration gene signature changes in CPI-naïve patient samples*
- *Spearman test was used for significance*
- *Anti-PD-1 datasets obtained from references 2, 5*

FLX475 monotherapy alters spatial organization and immunological phenotype in tumors

FLX475 monotherapy impacts CD8/Treg ratio and sub-localization of immune cells
Reduction of Treg cell migration from stroma to viable tumor regions

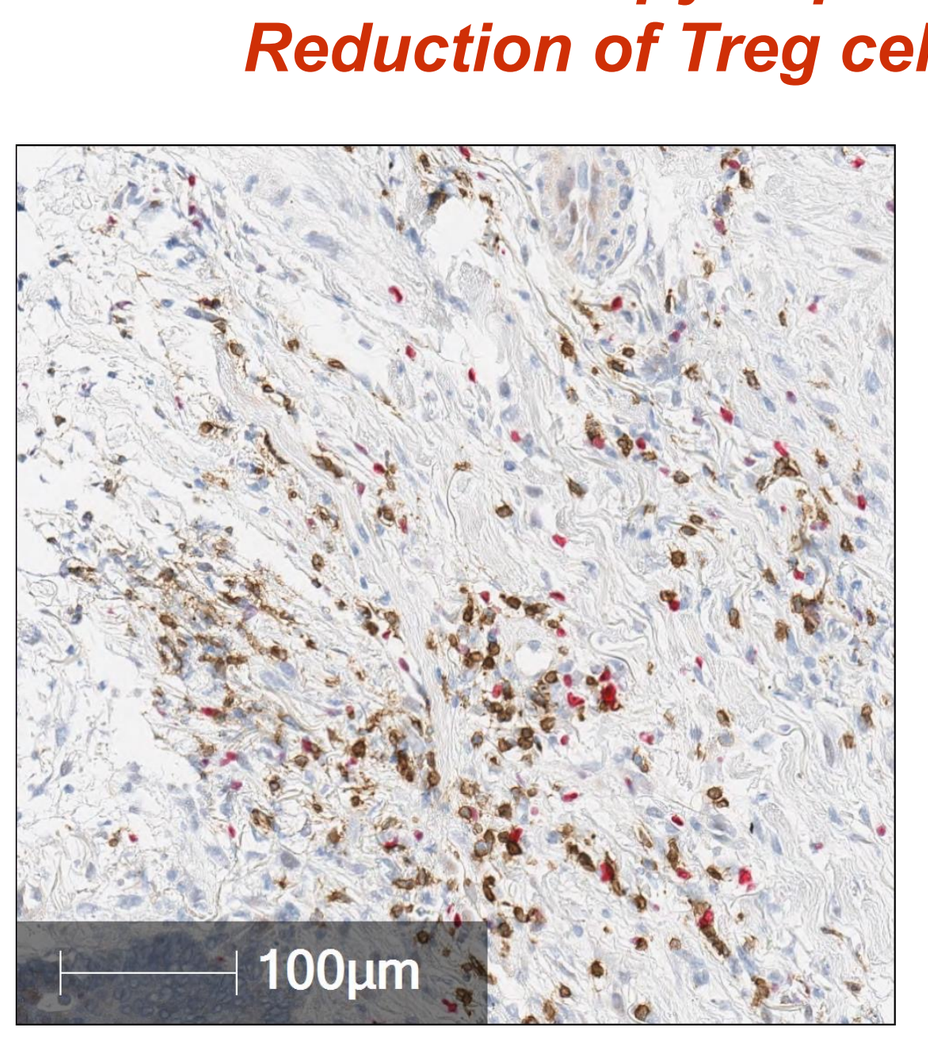
100μm

FOXP3 CD8

- *Representative staining derived from esophageal cancer patient*

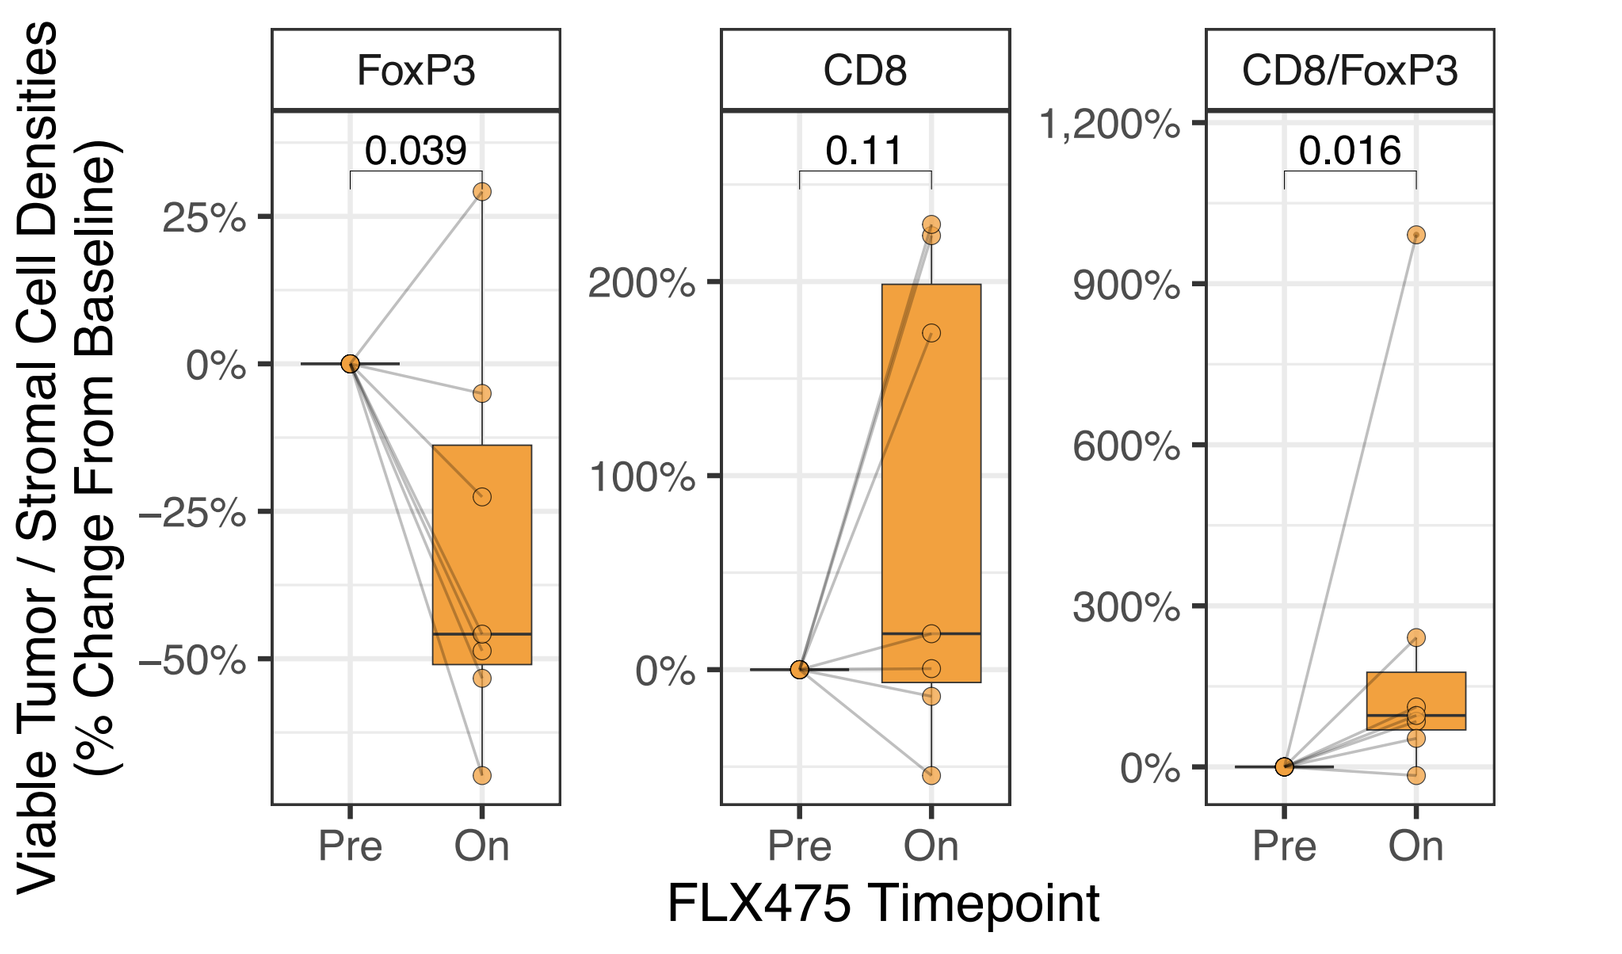
FoxP3
CD8
CD8/FoxP3
0.039
0.11
0.016
Viable Tumor / Stromal Cell Densities
(% Change From Baseline)
Pre
On
FLX475 Timepoint

- *One-tailed paired Wilcoxon test was used to evaluate significance*
- *HALO (Indica Labs, version 3.5) was used to quantify cell densities and proximities*

Spatial proximity between CD8+ and Treg are increased after FLX475 monotherapy

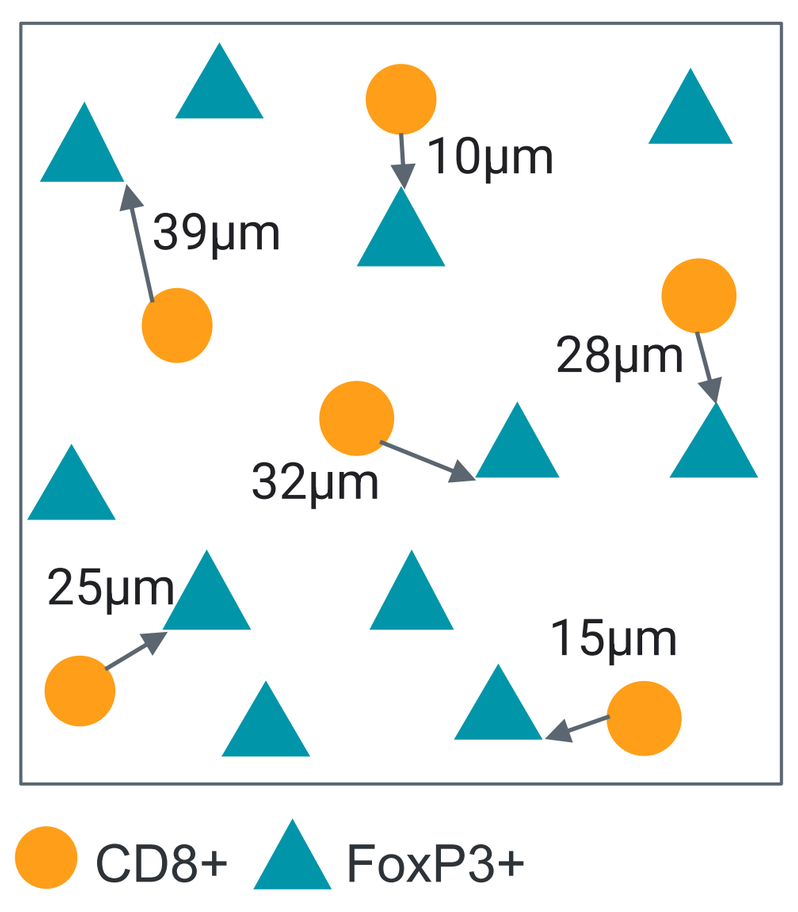
10μm
39μm
28μm
32μm
25μm
15μm
CD8+
FoxP3+

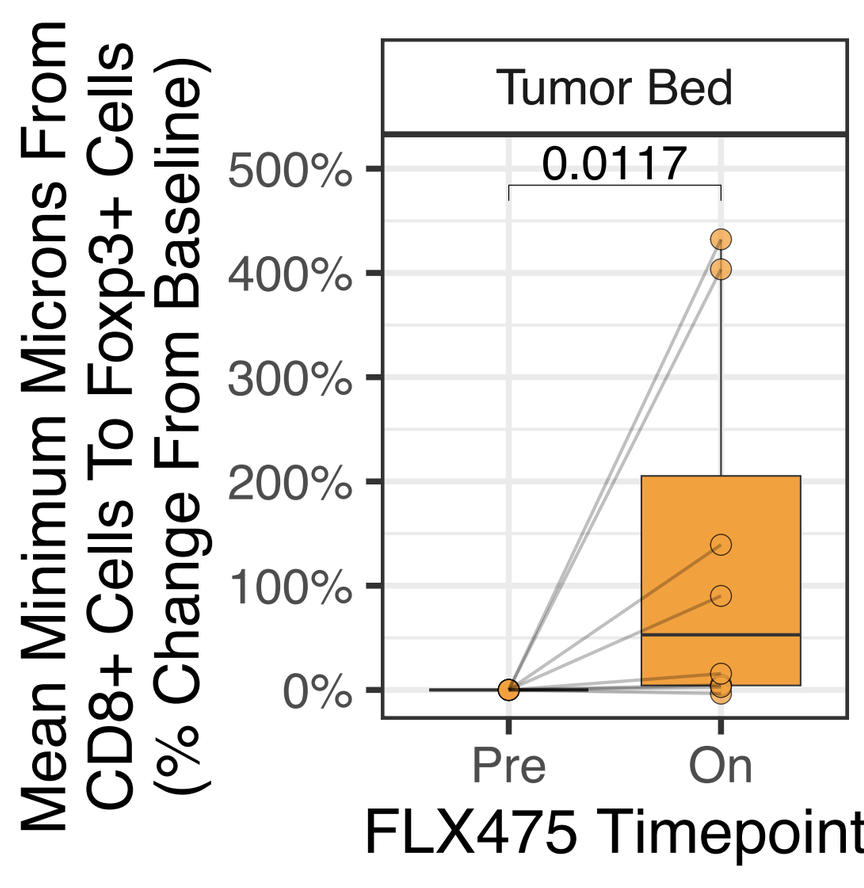
Tumor Bed
0.0117
Mean Minimum Microns From
CD8+ Cells To Foxp3+ Cells
(% Change From Baseline)
Pre
On
FLX475 Timepoint

- *One-tailed paired Wilcoxon test was used to evaluate significance*

FLX475 decreased Treg and increases pro-immunity associated gene signatures in the TME

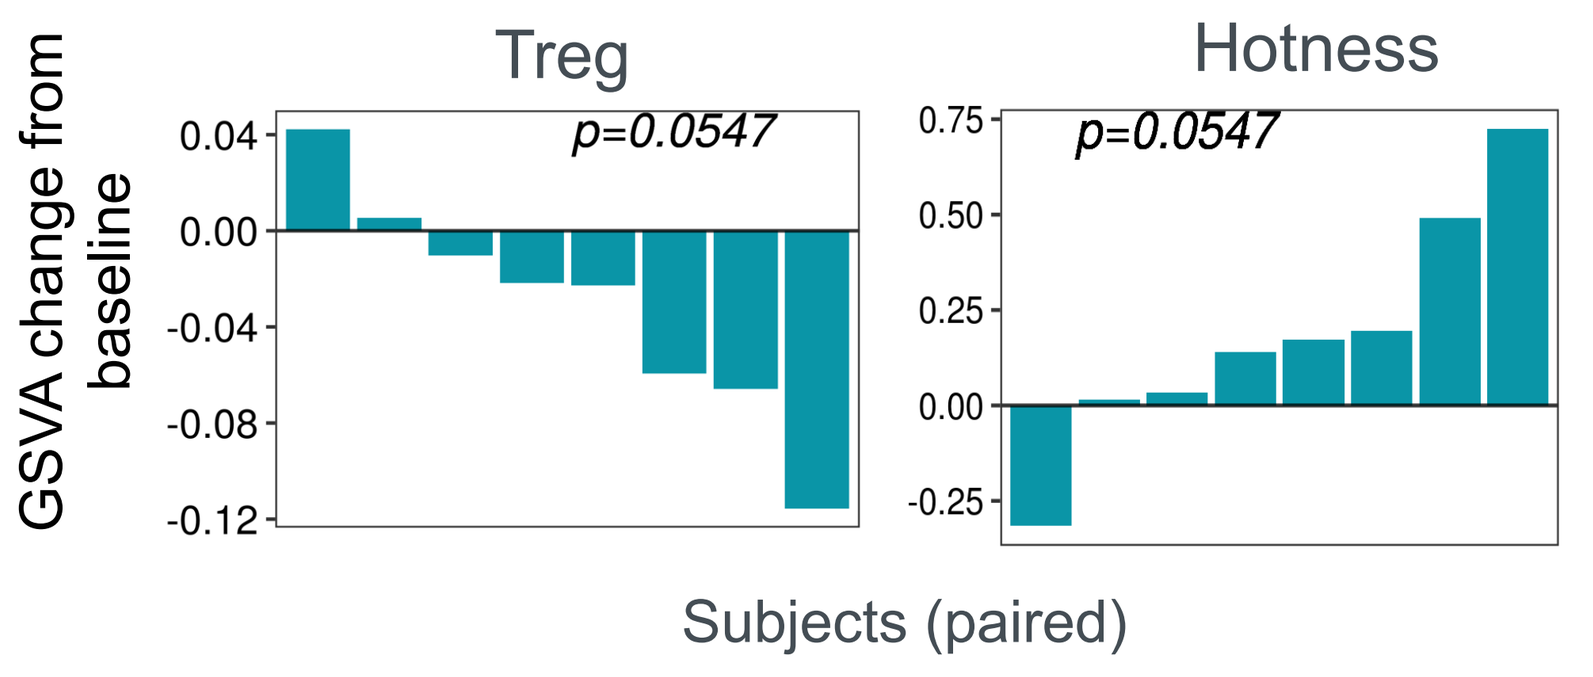
Treg
Hotness
p=0.0547
p=0.0547
GSVA change from
baseline
Subjects (paired)

- *Gene Set Variance Analysis (GSVA) was used to evaluate immune infiltration gene signature changes*
- *One-tailed paired Wilcoxon test was used for significance*

***Hotness includes CD8A, INFG, GZMB, PRF1*
***Treg includes FOXP3, CCL17, CCL22*

Baseline levels of intra-tumoral Treg has the potential to predict clinical benefit to FLX475/pembrolizumab and be used for patient stratification

FLX475/pembrolizumab predictive features in tumors are enriched for Treg signatures, which is not seen in anti-PD-1 monotherapy

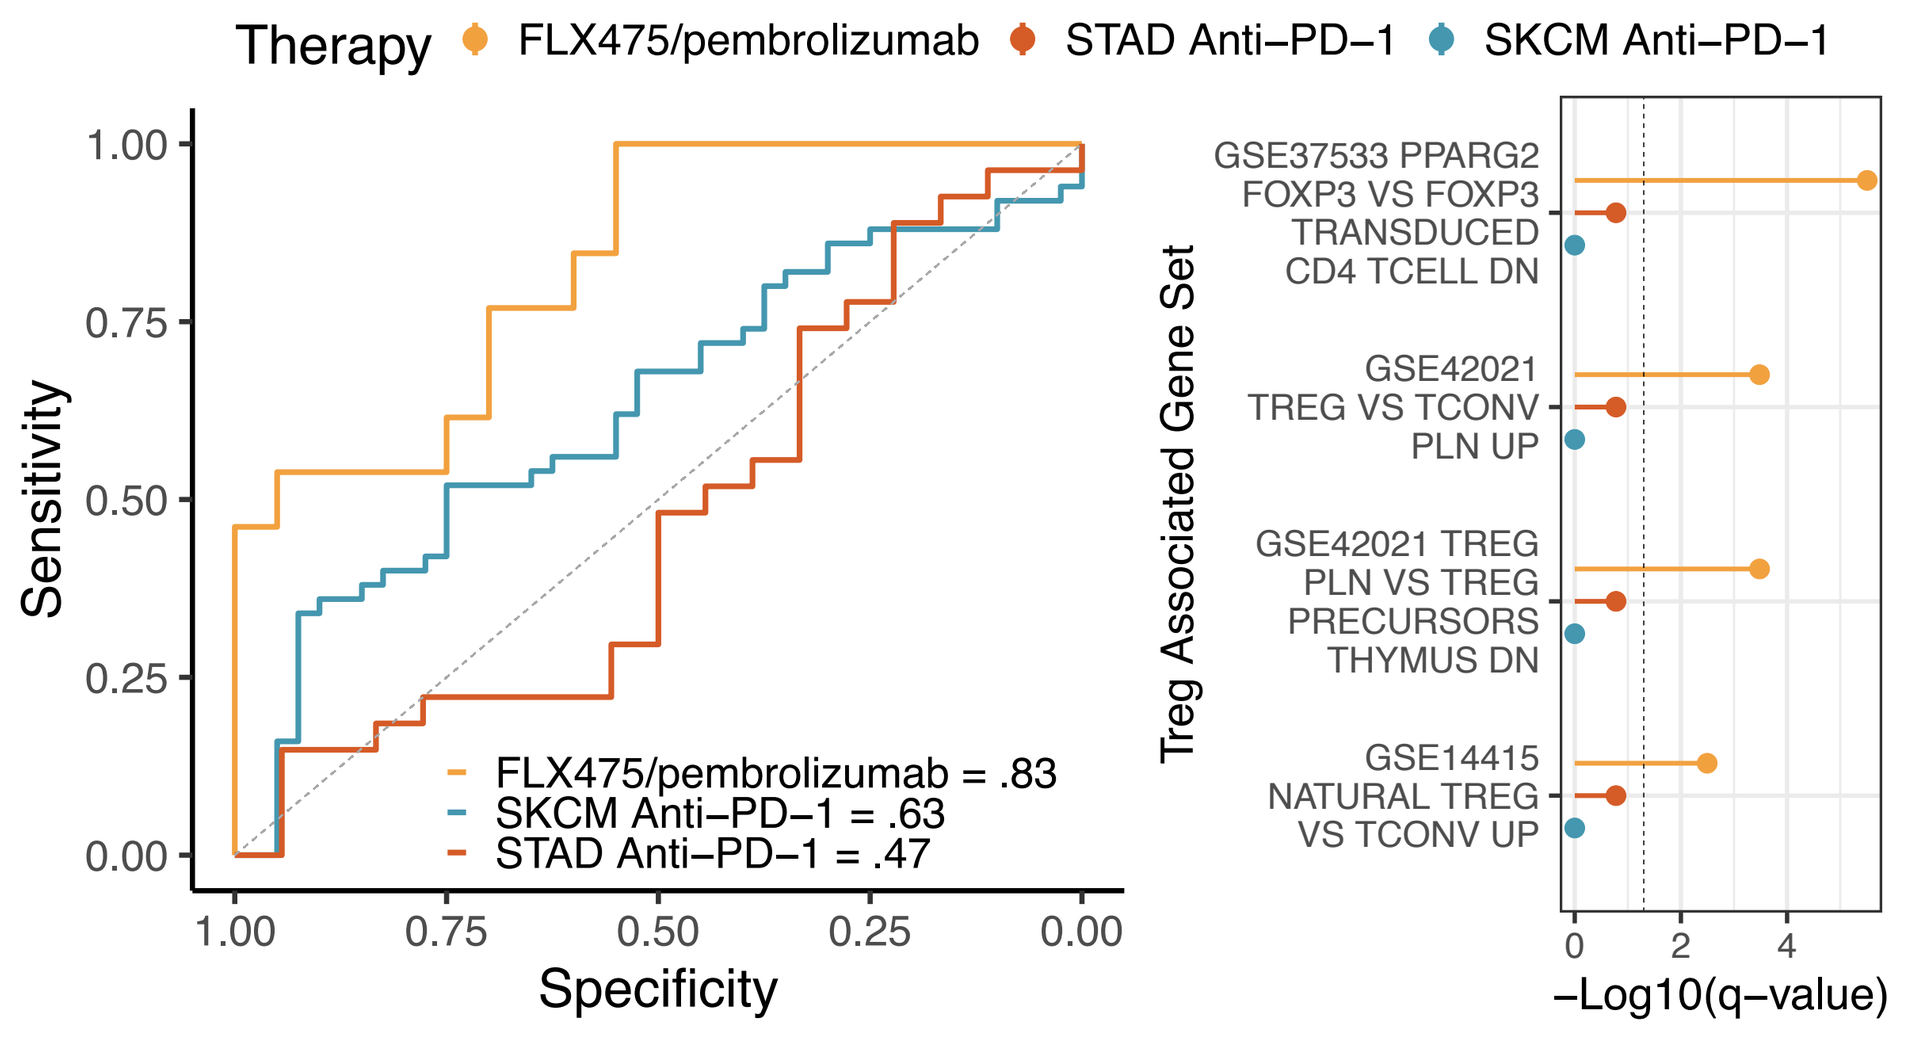
Therapy
FLX475/pembrolizumab
STAD Anti−PD−1
SKCM Anti−PD−1
Sensitivity
Specificity
FLX475/pembrolizumab = .83
SKCM Anti−PD−1 = .63
STAD Anti−PD−1 = .47
Treg Associated Gene Set
GSE37533 PPARG2
FOXP3 VS FOXP3
TRANSDUCED
CD4 TCELL DN
GSE42021
TREG VS TCONV
PLN UP
GSE42021 TREG
PLN VS TREG
PRECURSORS
THYMUS DN
GSE14415
NATURAL TREG
VS TCONV UP
−Log10(q−value)

- *An unbiased machine learning method was applied to identify gene expression features up-regulated in responders vs non-responders (hypergeometric test performed separately on each dataset)*
- *Points to the right of the dotted line have an adjusted p-value < 0.05*
- *Treg gene sets were selected from the MSigDB C7 immunologic signatures*

FLX475/pembrolizumab responders have higher intra-tumoral Tregs at baseline, not seen in anti-PD-1 monotherapy responders

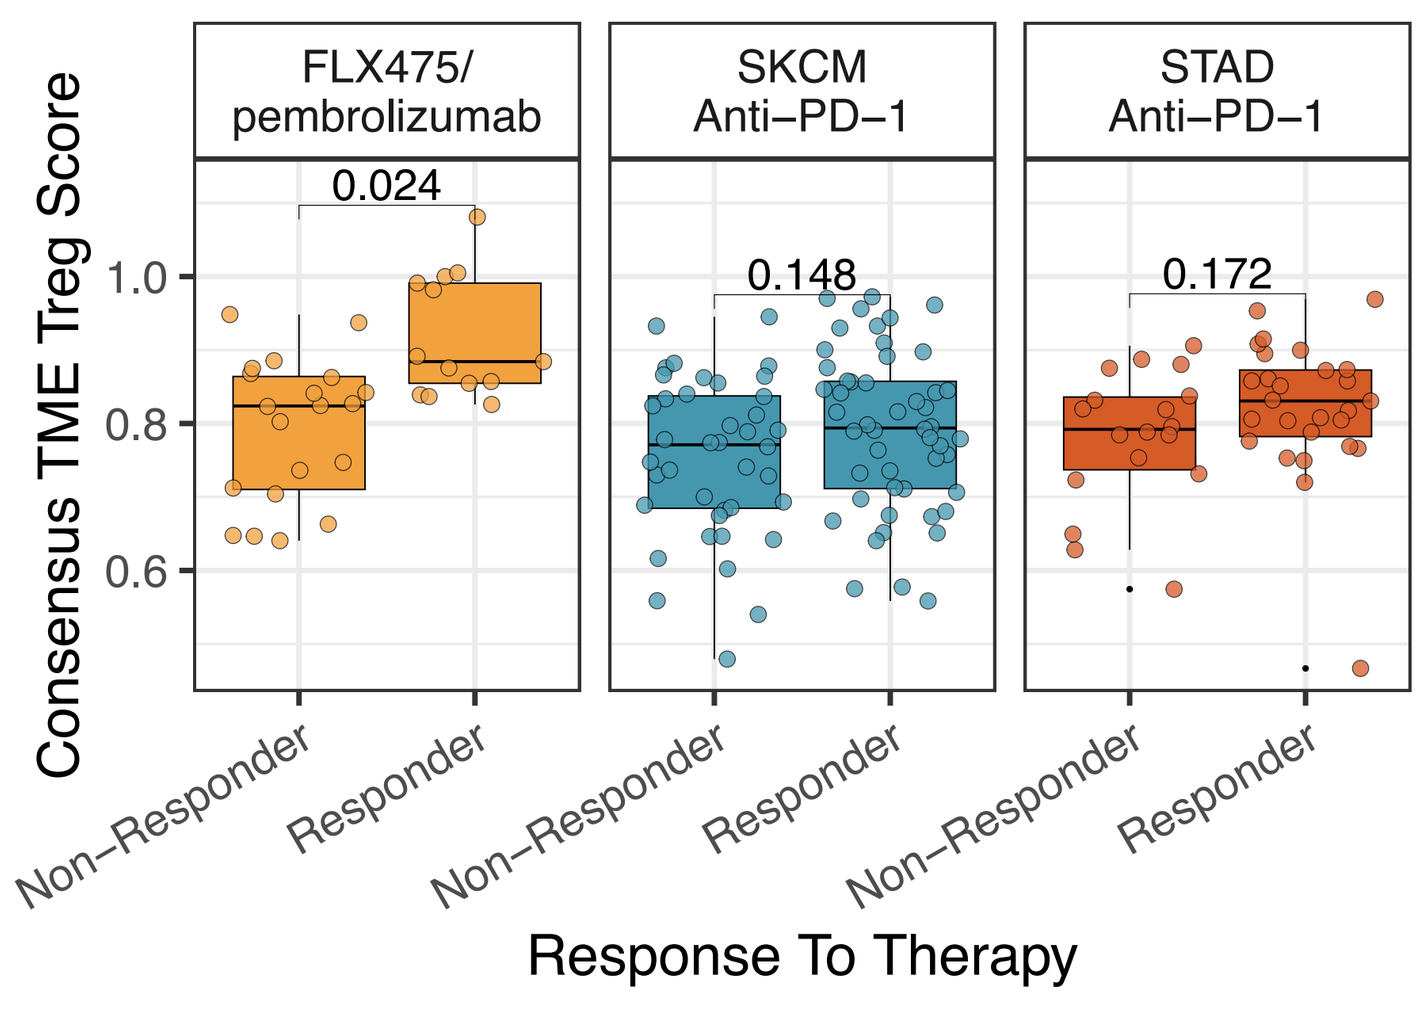
FLX475/
pembrolizumab
SKCM
Anti−PD−1
STAD
Anti−PD−1
0.024
0.148
0.172
Consensus TME Treg Score
Non−Responder
Responder
Response To Therapy

- *P-values were adjusted for cancer type and study*
- *Similar results were observed using Quantiseq and xCell immune deconvolution methods*
- *SKCM data from references 1,2 and 5; STAD data from reference 3*

Lower levels of peripheral Treg cells at baseline is associated with favorable response to FLX475/pembrolizumab

Progression free survival
All indications

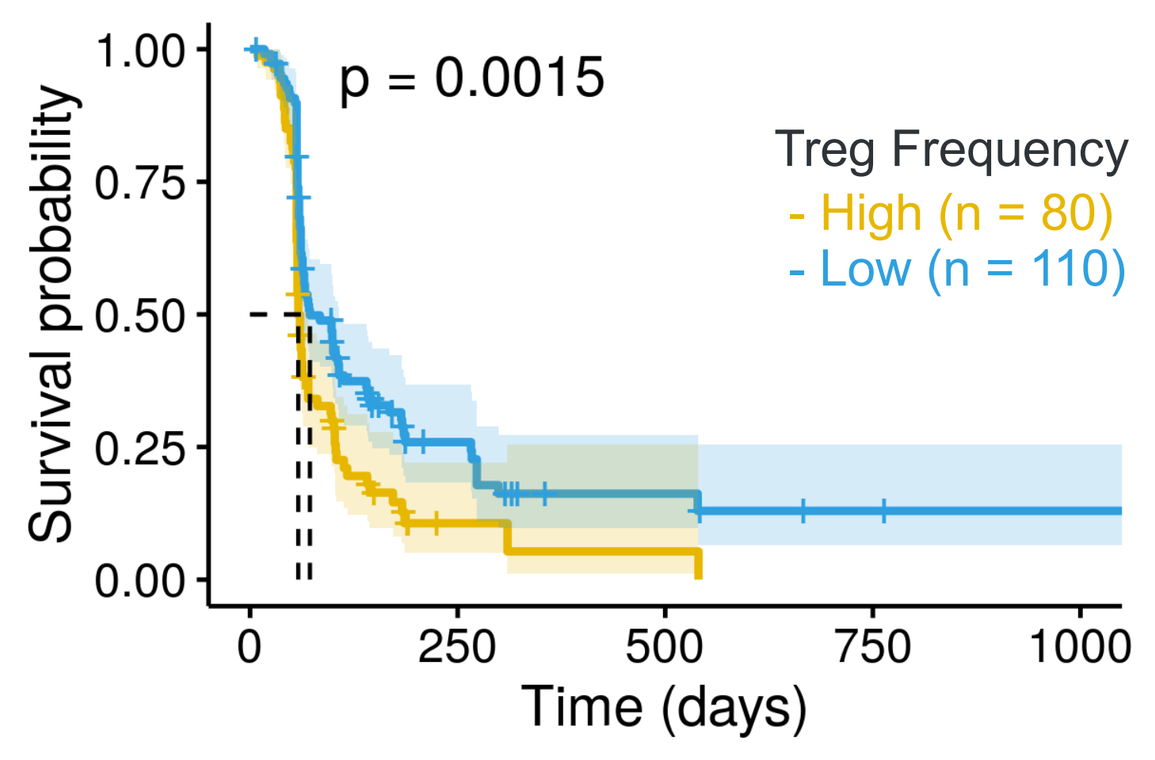
p = 0.0015
Treg Frequency
- High (n = 80)
- Low (n = 110)
Survival probability
Time (days)

Baseline peripheral levels of Treg cells by indication
Cohorts with higher number of patients are highlighted

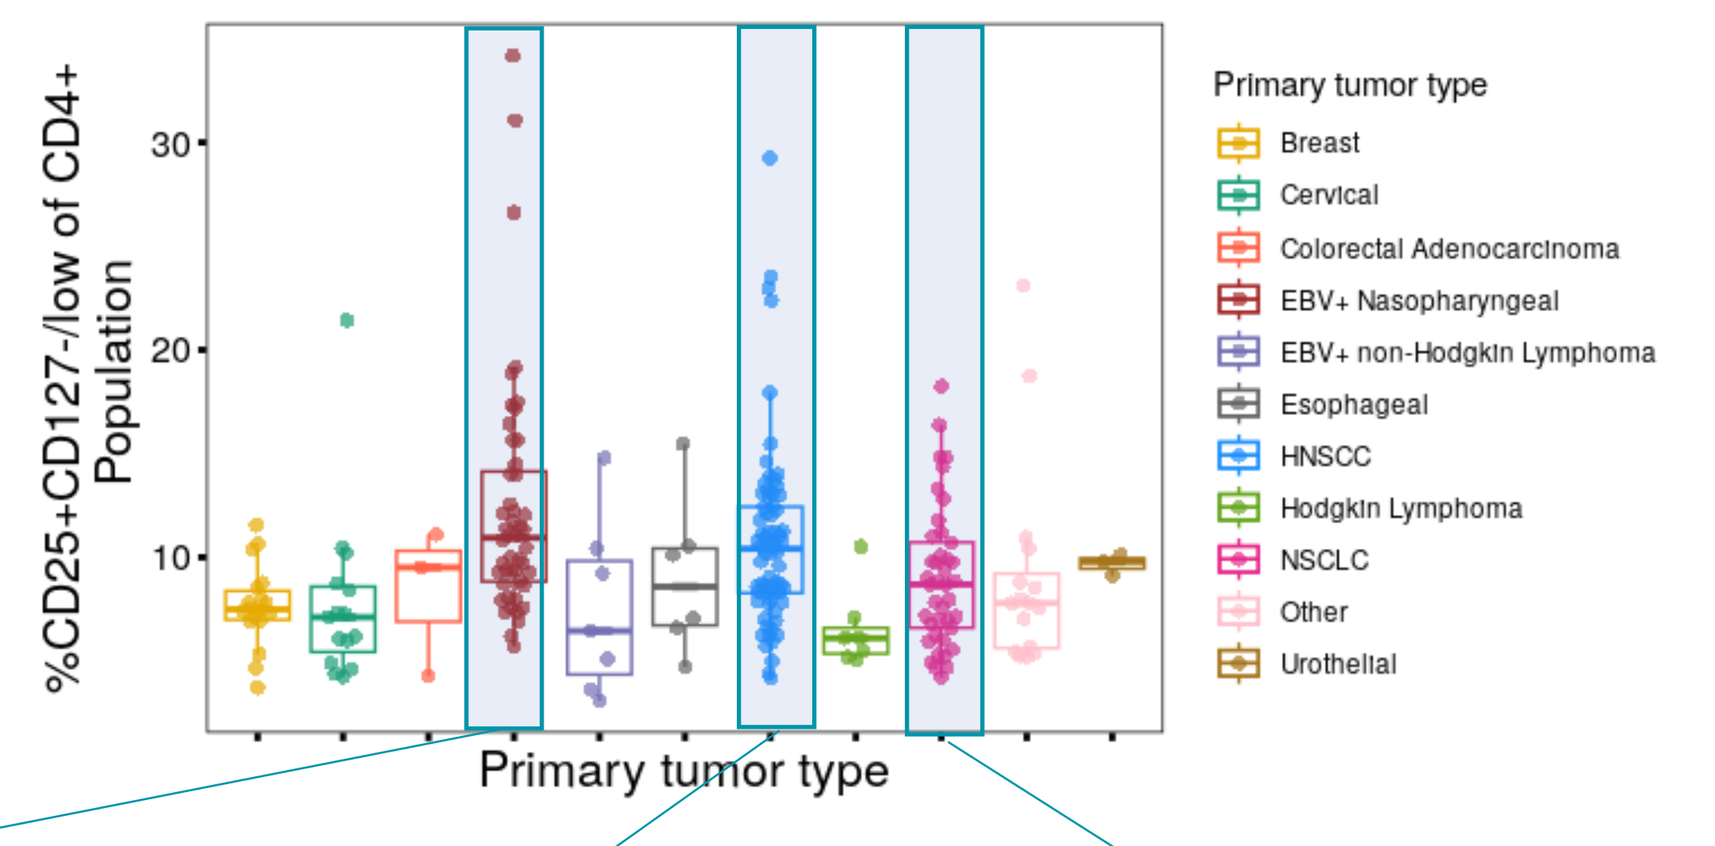
%CD25+CD127-/low of CD4+
Population
Primary tumor type
Primary tumor type
Breast
Cervical
Colorectal Adenocarcinoma
EBV+ Nasopharyngeal
EBV+ non-Hodgkin Lymphoma
Esophageal
HNSCC
Hodgkin Lymphoma
NSCLC
Other
Urothelial

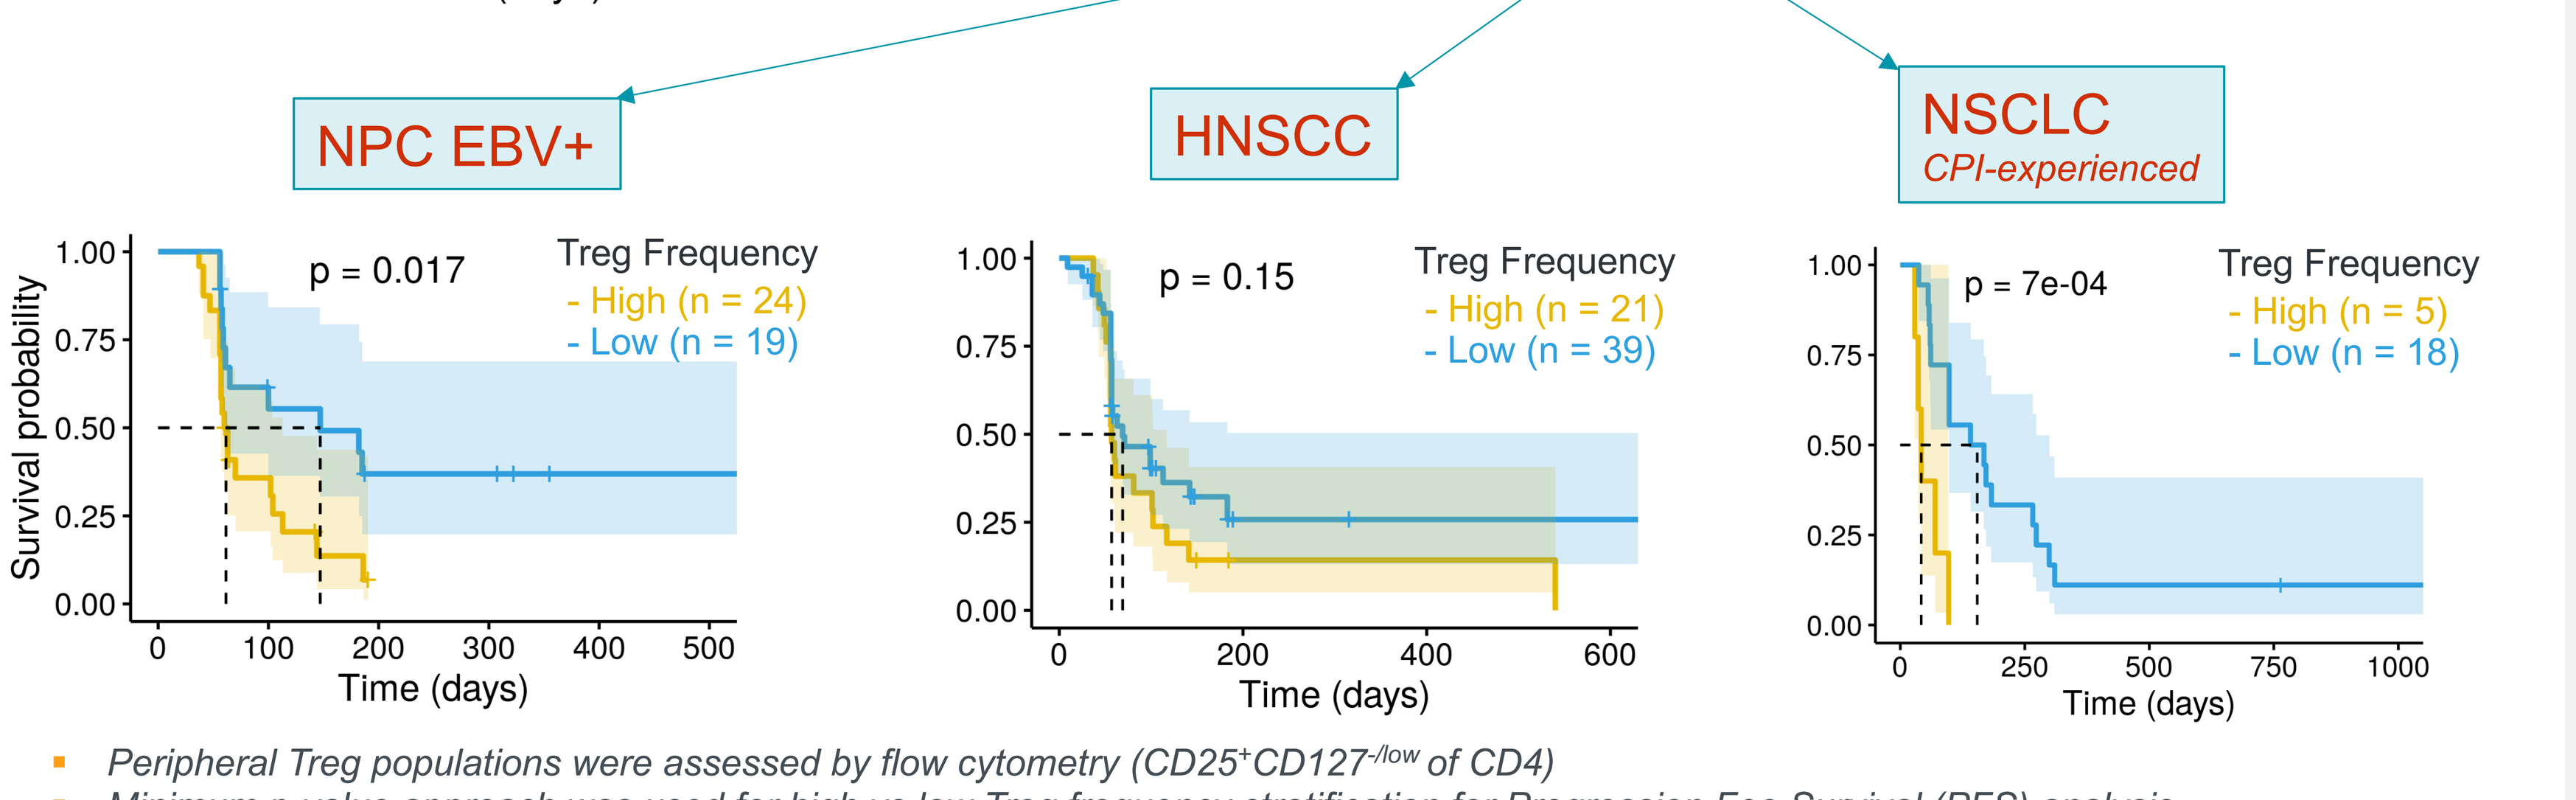
NPC EBV+
HNSCC
NSCLC
CPI-experienced
p = 0.017
Treg Frequency
- High (n = 24)
- Low (n = 19)
p = 0.15
Treg Frequency
- High (n = 21)
- Low (n = 39)
p = 7e-04
Treg Frequency
- High (n = 5)
- Low (n = 18)
Survival probability
Time (days)

- *Peripheral Treg populations were assessed by flow cytometry ($CD25^{+}CD127^{-/low}$ of CD4)*
- *Minimum p-value approach was used for high vs low Treg frequency stratification for Progression Fee Survival (PFS) analysis*
- *NSCLC includes CPI-experienced patients*
- *HNSCC includes both CPI-experienced and CPI-naïve patients*

Peripheral Treg cell accumulation upon FLX475 +/- pembrolizumab is a biomarker for biological activity of the drug

FLX475 +/- pembrolizumab increases Treg in a dose-dependent manner

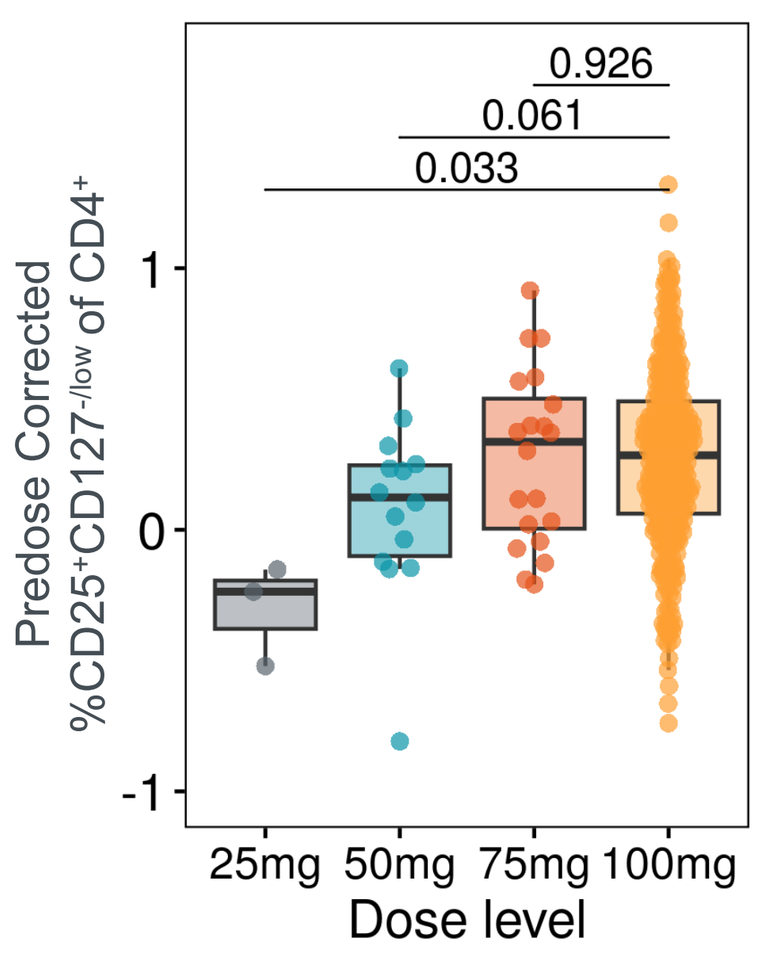
0.926
0.061
0.033
Predose Corrected
%CD25+CD127-/low of CD4+
25mg
50mg
75mg
100mg
Dose level

- *Change in Treg at C2D1 is shown*
- *Stats: two-sided t-test was used for pair-wise comparisons*

Peripheral Treg increase over time upon FLX475 +/- pembrolizumab

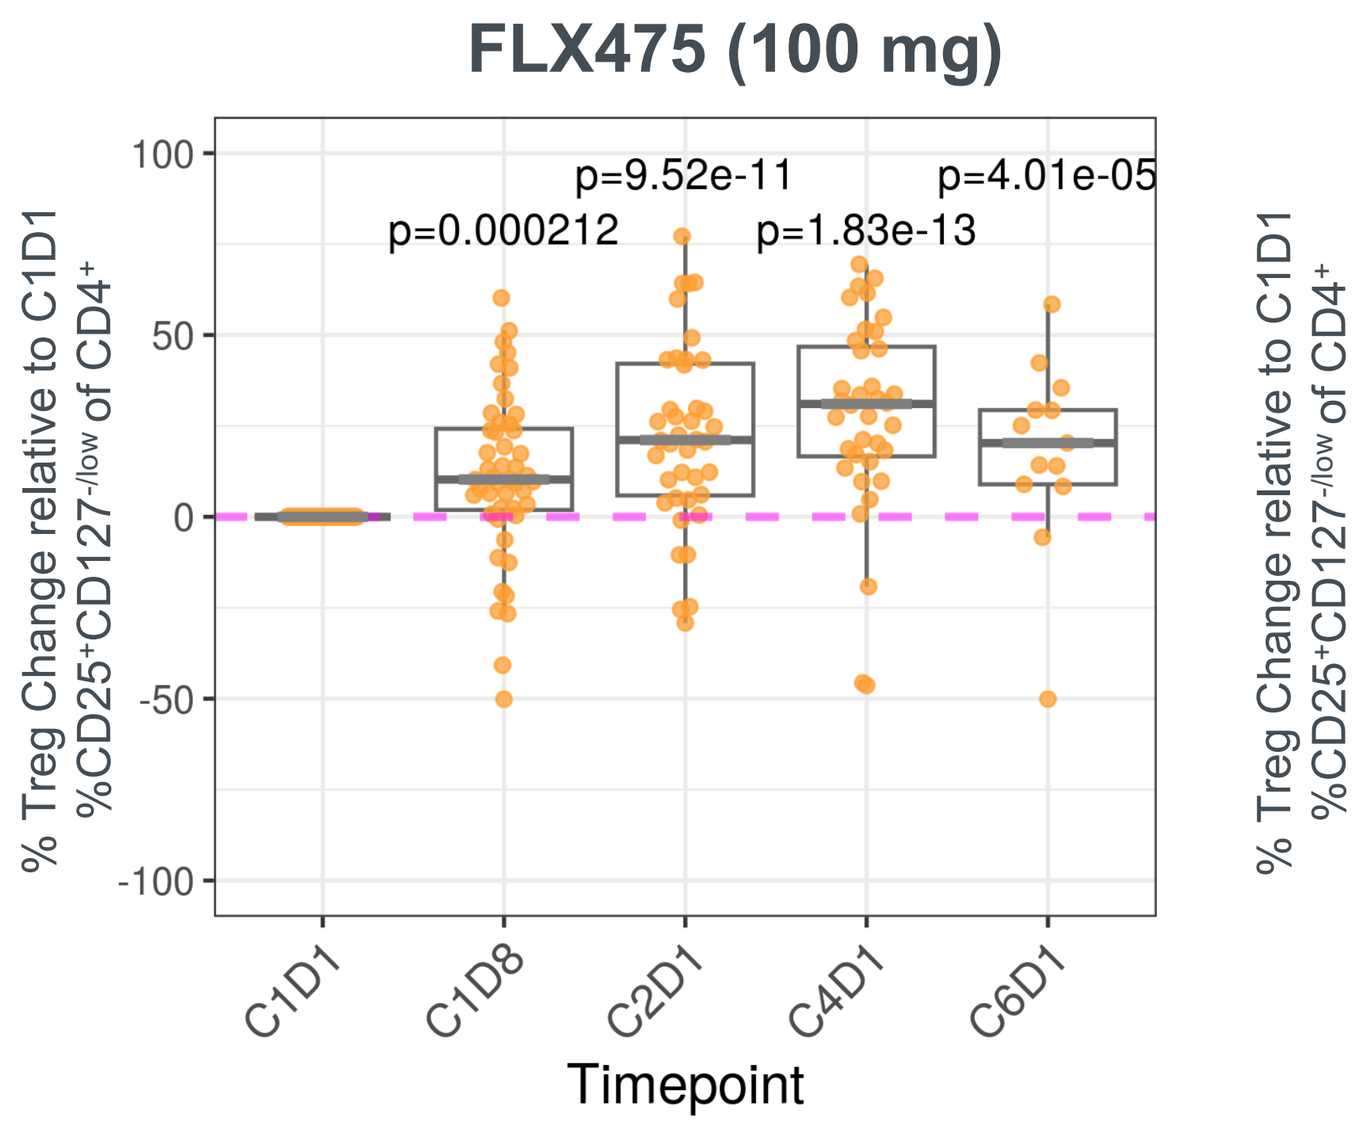
FLX475 (100 mg)
p=0.000212
p=9.52e-11
p=1.83e-13
p=4.01e-05
% Treg Change relative to C1D1
%CD25+CD127-/low of CD4+
C1D1
C1D8
C2D1
C4D1
C6D1
Timepoint

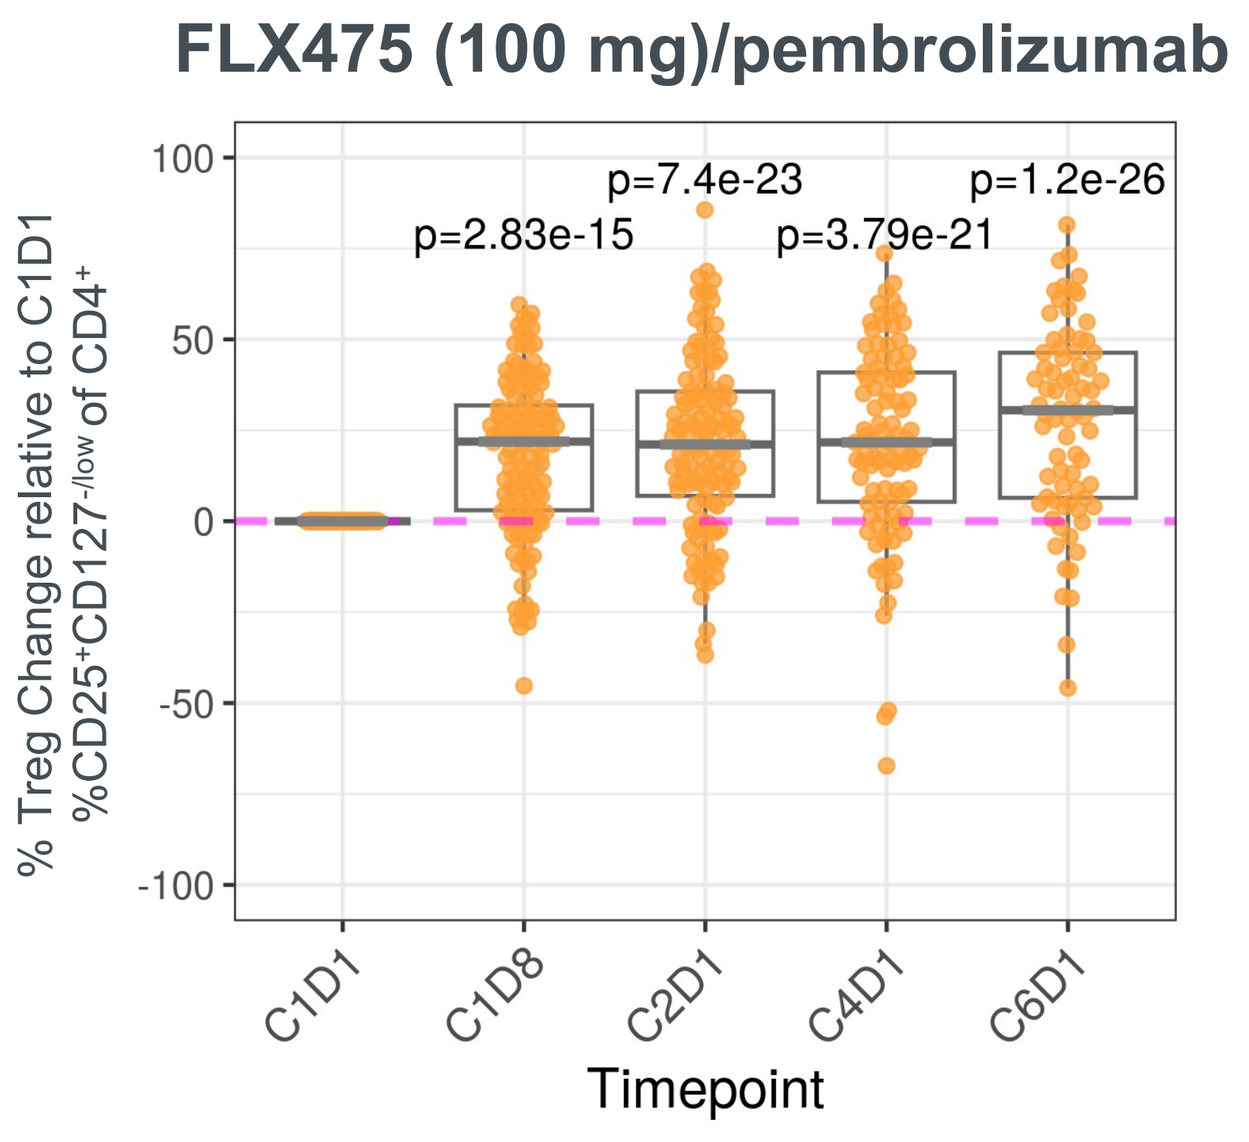
FLX475 (100 mg)/pembrolizumab
p=2.83e-15
p=7.4e-23
p=3.79e-21
p=1.2e-26
% Treg Change relative to C1D1
%CD25+CD127-/low of CD4+
C1D1
C1D8
C2D1
C4D1
C6D1
Timepoint

- *Cross bars represent median of the points across visits*
- *1 outlier at C1D8 below -100 in the FLX475/pembrolizumab group is outside of the vertical boundary of the graph*
- *Linear mixed effect model was used to test statistical significance*
- *Purple dashed line marks 25% change for visualization purposes*

Published anti-PD-(L)-1 RNA-seq datasets used for comparative analyses with FLX475 +/- pembrolizumab

Study	Drug	Cancer Type	Year	PMID
[1]Hugo et al.	Pembrolizumab	SKCM (Skin Cutaneous Melanoma)	2016	26997480
[2]Riaz et al.	Nivolumab	SKCM (Skin Cutaneous Melanoma)	2017	29033130
[3]Kim et al.	Pembrolizumab	STAD (Stomach Adenocarcinoma)	2018	30013197
[4]Mariathasan et al.	Atezolizumab	BLCA (Bladder Urothelial Carcinoma)	2018	29443960
[5]Gide et al.	Pembrolizumab, Nivolumab	SKCM (Skin Cutaneous Melanoma)	2019	30753825

- All data sets were processed using the same RNA-seq pipeline to reduce batch effects

Conclusions

- FLX475 monotherapy shows biological activity in patients, demonstrated by alteration in intra-tumoral immunophenotype, spatial distancing of immune cells and upregulation of gene signatures associated with anti-tumor immunity and anti-PD-1/L1 response.
- FLX475 monotherapy or in combination with pembrolizumab leads to a significant increase in frequency of Treg cells in the blood, acting as a pharmacodynamic biomarker for drug activity.
- Comparative analyses with published anti-PD-1 RNAseq datasets revealed that intratumoral CCR4 and its ligands (CCL17/CCL22) are upregulated in CPI-naïve patients upon anti-PD-1 treatment, suggesting increased Treg cell trafficking. On the contrary, FLX475 limited CCR4-associated Treg accumulation in the TME of patients receiving FLX475/pembrolizumab combination.
- In contrast to anti-PD-1 monotherapy, FLX475/pembrolizumab responders have significantly higher intra-tumoral Tregs relative to non-responders at baseline. This suggests that patient populations with enriched intratumoral Tregs may benefit from FLX475/pembrolizumab as compared to anti-PD-1 alone.
- In peripheral blood, lower levels of Treg cells at baseline correlate with favorable response and better PFS for patients treated with FLX475/pembrolizumab.

Acknowledgments: We would like to thank the patients, caregivers, clinical investigators and their staff for participation in our clinical trial. Thanks to our colleagues at RAPT Therapeutics and scientific advisors for helpful suggestions and discussion.
This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA