Discovery Of Potent And Selective Inhibitors Of USP7 With Anti-Tumor Activity In Vitro And In Vivo

Yamini Ohol, Michael Sun, Gene Cutler, Paul Leger, Dennis Hu, Berenger Biannic, Payal Rana, Cynthia Cho, Scott Jacobson, Steve Wong, Jerick Sanchez, Xinping Han, Kyle Young, Akinori Okano, Nick Shah, Betty Abraham, Deepa Pookot, Jack Maung, Delia Bradford, Nathan Kozon, Silpa Suthram, Lavanya Adusumilli, Deepika Kaveri, Oezcan Talay, Christophe Colas, Andrea Kim, Jacob Schwarz, David Wustrow, Dirk Brockstedt, Paul Kassner; **FLX Bio, Inc. South San Francisco, CA**

Abstract

USP7 is a deubiquitinase that regulates the levels of multiple downstream targets with roles in cancer progression and immune response. Inhibitors of USP7 (USP7i) may decrease oncogene function, increase tumor suppressor function, enhance immune function and sensitize tumor cells to DNA damaging agents. We have developed USP7i that are potent and selective in biochemical and cellular assays. Our USP7i reduce the viability of multiple p53-wild type cell lines, including blood cancer and neuroblastoma cell lines, as well as a subset of p53-mutant tumor cell lines in vitro. Further, oral administration of our USP7i inhibits MM.1S (multiple myeloma; p53-wild type) and H526 (small cell lung cancer; p53-mutant) tumor growth in vivo.

Multiple Beneficial Mechanisms of USP7 Inhibitors

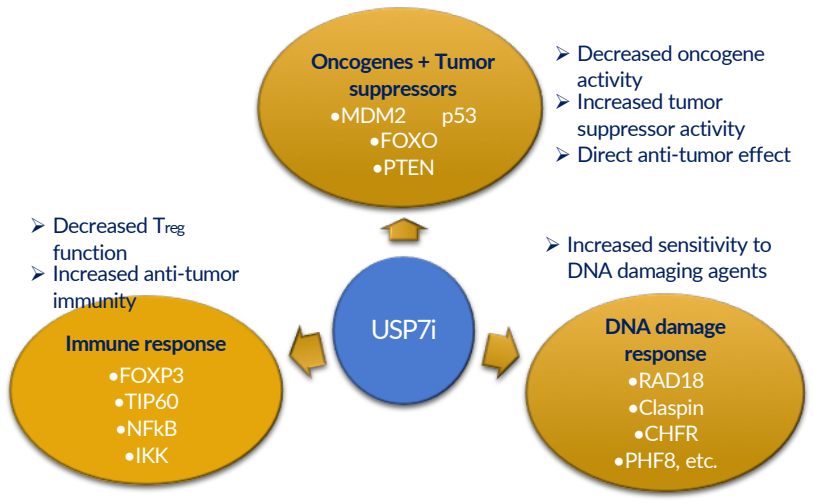
Oncogenes + Tumor suppressors
•MDM2 p53
•FOXO
•PTEN
Decreased oncogene activity
Increased tumor suppressor activity
Direct anti-tumor effect
Decreased Treg function
Increased anti-tumor immunity
Immune response
•FOXP3
•TIP60
•NFkB
•IKK
USP7i
Increased sensitivity to DNA damaging agents
DNA damage response
•RAD18
•Claspin
•CHFR
•PHF8, etc.

Biochemical Potency of Compounds Used in These Studies

USP7i	USP7 IC_{50}
FLX-A	10 nM
FLX-B	< 1 nM
FLX-C	< 1 nM
Active enantiomer	< 1 nM
Inactive enantiomer	< 100 nM
FLX-D (in vivo tool)	< 1 nM
FLX-E (in vivo tool)	< 1 nM

1 USP7 Inhibitors are Highly Selective and Potent in Biochemical and Cellular Assays

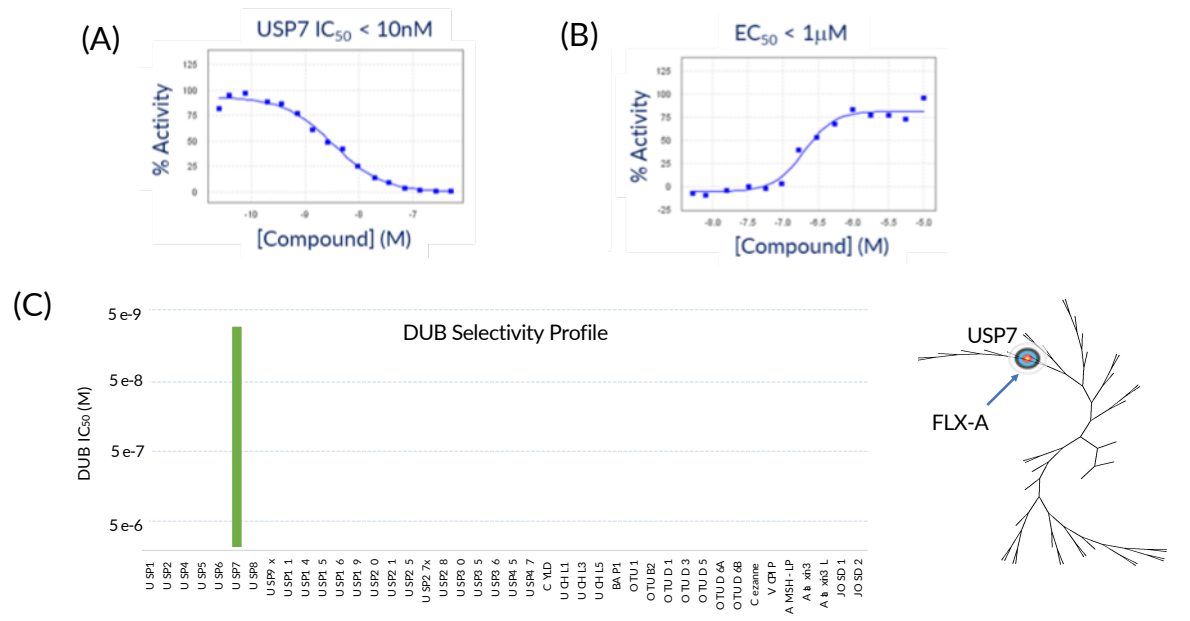
(A)
USP7 IC_{50} < 10nM
% Activity
[Compound] (M)
(B)
EC_{50} < 1µM
% Activity
[Compound] (M)
(C)
DUB Selectivity Profile
DUB IC_{50} (M)
USP7
FLX-A

(**A**) USP7 inhibitor inhibits USP7 in biochemical assay using rhodamine-labeled ubiquitin. (**B**) USP7 inhibitor elevates p53 in a luciferase reporter gene cell-based assay. (**C**) USP7 inhibitor IC_{50} is <10 nM for USP7 but >10 µM for all other DUBs tested.

2 USP7 Inhibitors Inhibit p53-Wild Type Tumors In Vitro and In Vivo

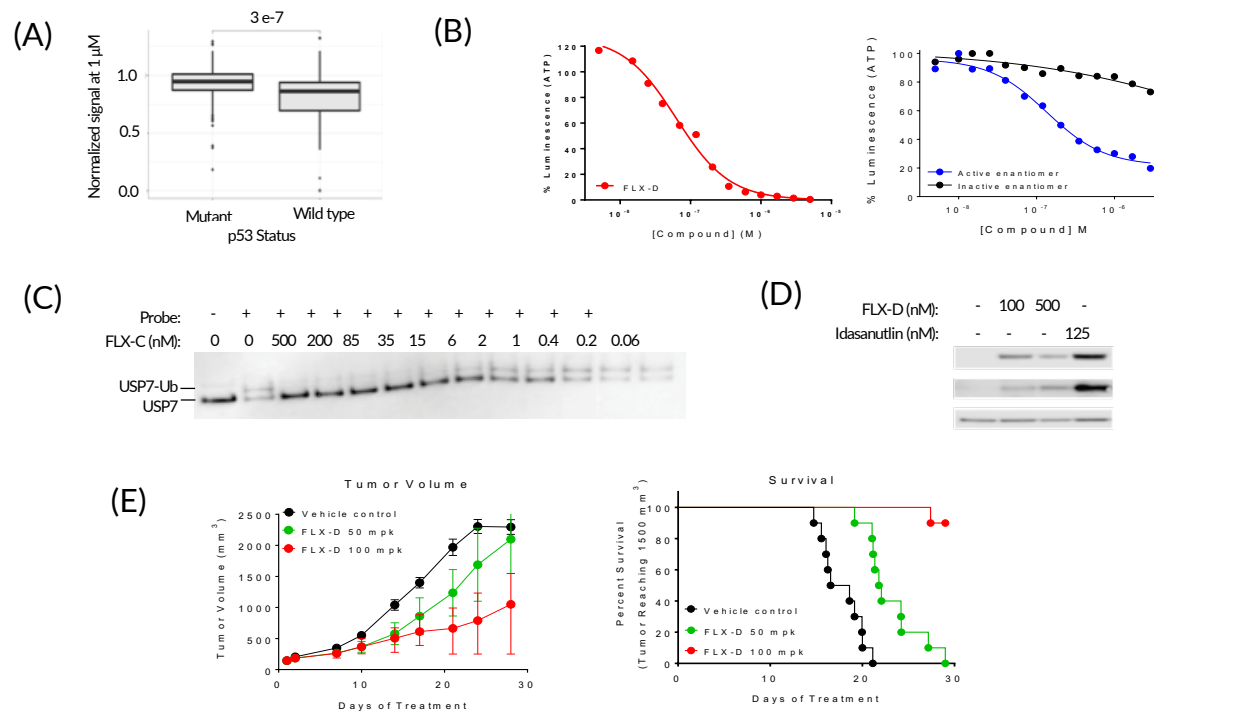
(A)
3 e-7
Normalized signal at 1 µM
Mutant
Wild type
p53 Status
(B)
FLX-D
Active enantiomer
Inactive enantiomer
[Compound] M
(C)
Probe:
FLX-C (nM): 0 0 500 200 85 35 15 6 2 1 0.4 0.2 0.06
USP7-Ub
USP7
(D)
FLX-D (nM): - 100 500 -
Idasanutlin (nM): - - - 125
(E)
Tumor Volume
Vehicle control
FLX-D 50 mpk
FLX-D 100 mpk
Days of Treatment
Survival

(**A**) p53 wild type status is a predictor of USP7i sensitivity. (**B**) MM.1S cells were treated with USP7 inhibitors for 5 days, after which viability was measured by CellTiter Glo assay. (**C**) MM.1S cells were treated with USP7 inhibitor for 4h, after which USP7 engagement was measured using Ub-PA probe. (**D**) MM.1S cells were treated with USP7 inhibitor or Idasanutlin for 6h, after which p53 and p21 levels were measured by Western blot. (**E**) NOD-scid mice engrafted with multiple myeloma MM.1S tumors were treated with USP7 inhibitor for the indicated time, and tumor volume and survival were monitored.

3 USP7 Inhibitors Inhibit p53-Mutant Tumors In Vitro and In Vivo

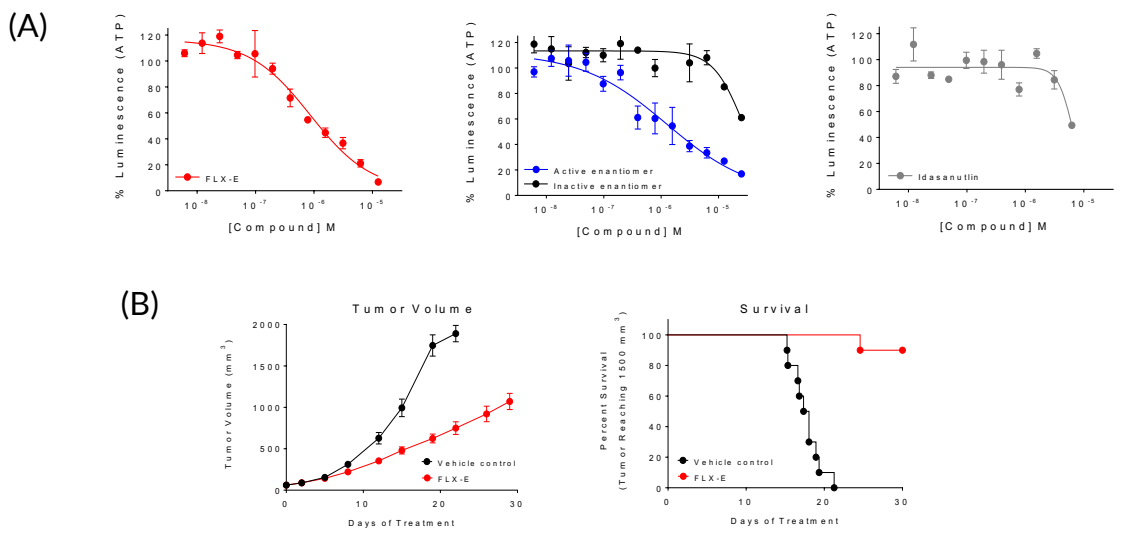
(A)
FLX-E
Active enantiomer
Inactive enantiomer
Idasanutlin
[Compound] M
(B)
Tumor Volume
Survival
Vehicle control
FLX-E
Days of Treatment

(**A**) H526 small cell lung cancer cells were treated with USP7 inhibitors or idasanutlin for 5 days, after which viability was measured by CellTiter Glo assay. (**B**) Nu/Nu mice engrafted with H526 tumors were treated with USP7 inhibitor for the indicated time, and tumor volume and survival were monitored.

4 USP7 Inhibitors Synergize With DNA-Damaging Agents, PIM and PI3K Inhibitors

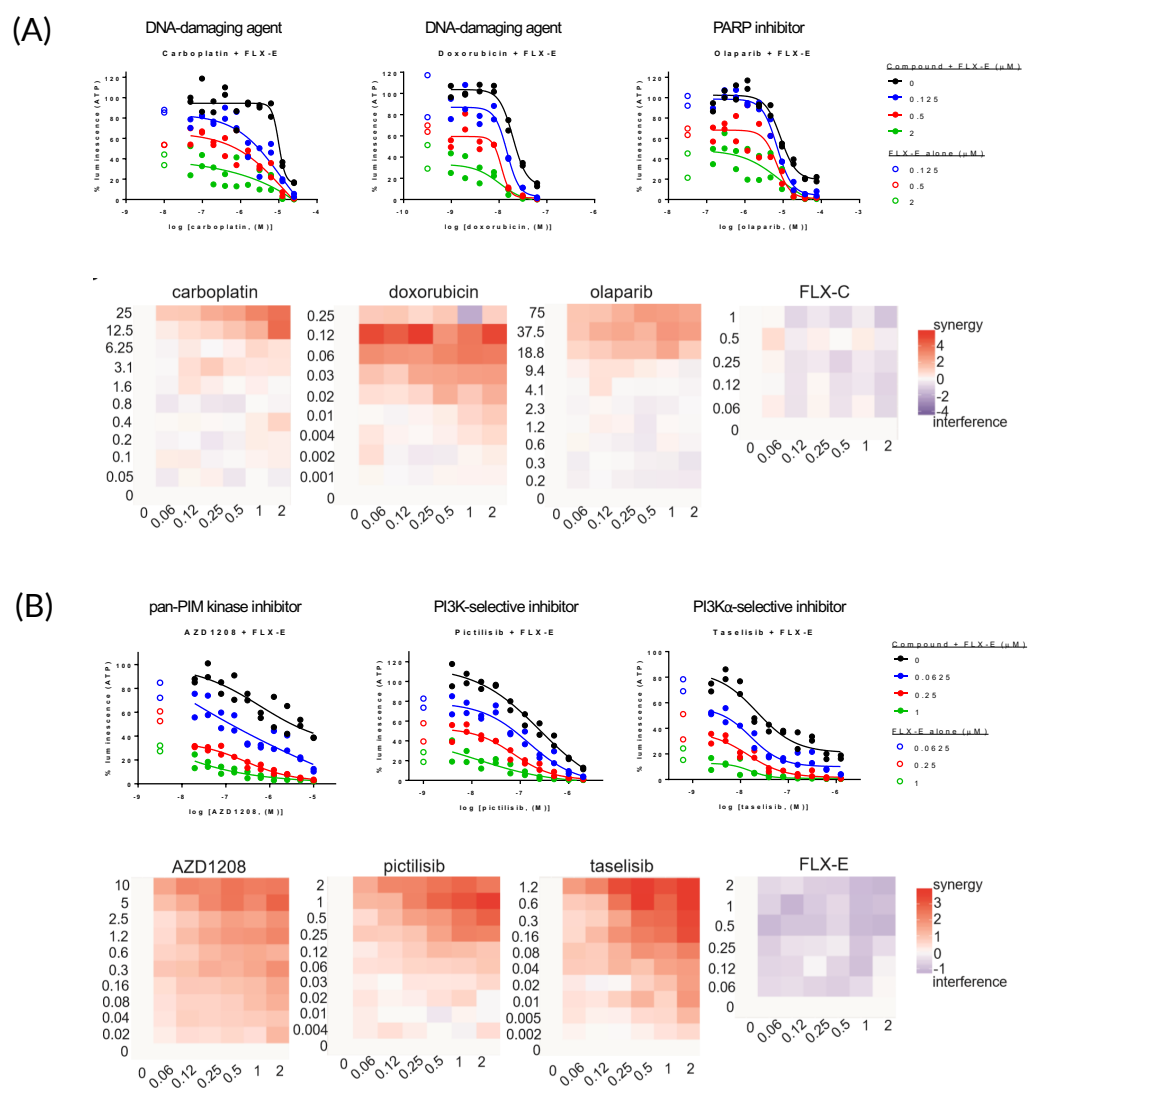
(A)
DNA-damaging agent
DNA-damaging agent
PARP inhibitor
carboplatin
doxorubicin
olaparib
FLX-C
synergy
interference
(B)
pan-PIM kinase inhibitor
PI3K-selective inhibitor
PI3Kα-selective inhibitor
AZD1208
pictilisib
taselisib
FLX-E

(**A**, **B**) H526 cells were treated with indicated drug (y-axis) and USP7 inhibitor (x-axis) at indicated dose (µM) for 5 days, after which viability was measured by CellTiter Glo assay and synergy was calculated by Bliss analysis. Combinations of USP7 inhibitors with DNA-damaging agents and PARP inhibitor were tested in (**A**), PIM and PI3K inhibitors in (**B**).

Conclusions

- We have developed a novel series of potent, highly selective, orally available small molecule USP7 inhibitors with anti-tumor activity in vitro and in vivo.
- FLX USP7 inhibitors are effective against multiple blood cancer and *MYCN*-amplified neuroblastoma cell lines with p53-WT status in vitro.
- FLX USP7 inhibitors are effective against a subset of p53-mutant tumor cell lines and synergize with DNA-damaging agents, PIM and PI3K inhibitors.
- Oral delivery of FLX USP inhibitors reduces growth of MM.1S (multiple myeloma; p53-wild type) and H526 (small cell lung cancer; p53-mutant) tumors and prolongs survival in vivo.

DO NOT POST